Secretory component: an universal microbial scavenger at mucosal surfaces?

Clémentine Perrier[1,2], Norbert Sprenger[2] and Blaise Corthésy[1]

[1] Service d'immunologie et d'allergie, R & D laboratory, CHUV, Rue du Bugnon 46, 1011 Lausanne
[2] Nestlé Research Centre, Vers-chez-les-Blanc, 1000 Lausanne 26

Introduction and Aims

What is secretory component ?

Secretory component (SC) is a member of the immunoglobulin superfamily and is secreted at mucosal surfaces (i.e. gastrointestinal, respiratory and urogenital tracts). SC is derived from the polymeric immunoglobulin receptor (pIgR) which transports IgA across the epithelium. SC can be found in mucosal secretions either in association with secretory IgA (SIgA) or alone as free SC.

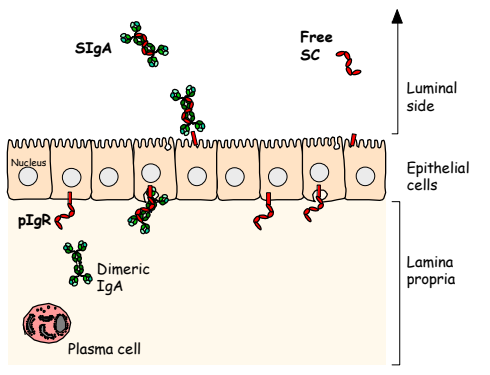
SIgA
Free SC
Luminal side
Epithelial cells
Lamina propria
Nucleus
pIgR
Dimeric IgA
Plasma cell

Schematic representation of the transport of secretory IgA across the epithelium (left). Dimeric IgA is produced by plasma cells in the lamina propria and binds subsequently to pIgR expressed at the basolateral surface of epithelial cells. The complex is then transcytosed to the luminal side and released as SIgA through proteolitic cleavage. pIgR can also be transported as such, leading to the release of free SC at mucosal surfaces (right).

SC is a highly glycosylated protein:

15-20 % of the total molecular mass of SC are carbohydrates. The composition and structure of these sugars suggests a biological role as scavenger for SC.

Aims:

Evaluate the possible role of SC as microbial scavenger of different pathogenic agents, and determine the role of sugars in this process.

Model, Proteins and Pathogenic agents

Model:

As a mimic of the intestinal epithelium, polarized human colonic adenocarcinoma cells (Caco-2) are grown on transwell filters.

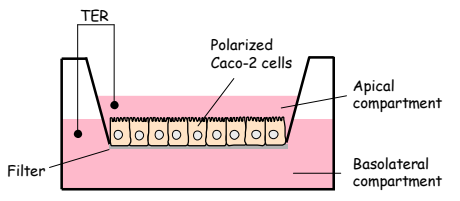
TER
Polarized Caco-2 cells
Apical compartment
Basolateral compartment
Filter

In vitro model of intestinal epithelium using Caco-2 cells grown on transwell filters. A Trans-epithelial resistance (TER) can be measured between apical and basolateral compartments when cells are differentiated.

Proteins:

Two different sources of SC were used and compared:

- **Human colostral SC (SCcol)**
 Purification procedure includes a combination of gel filtration and ion-exchange chromatography.
- **Human recombinant SC (SCrec)**
 Production is achieved in Chinese hamster ovary (CHO) cells and purified by gel filtration.

A major interest of this work is to determine the biological contribution of carbohydrates of SC. Therefore, **deglycosilated SC (SCdg)** was prepared using a N-glycosidase F.

Pathogenic agents:

Bacterial components and a viral pathogen were studied:

- the toxin A from *Clostridium difficile.*
- Intimin from enteropathogenic *Escherichia coli.*
- Rotavirus.

Preliminary Results

SC protects epithelial cells from cytotoxic effects of toxin A

Toxin A binds to epithelial cells and destructs the monolayer, leading to a drop of the transepithelial resistance (TER). When toxin A is preincubated with SC before apical addition to the cell monolayer, the TER is not altered.

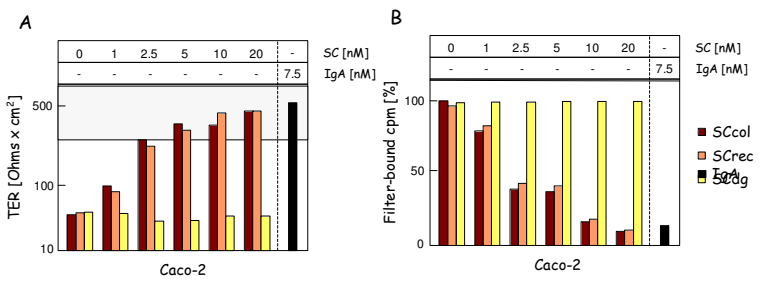
A
B
SC [nM]
IgA [nM]
TER [Ohms x cm²]
Filter-bound cpm [%]
Caco-2
SCcol
SCrec
SCdg
IgA

Graph representing the transepithelial resistance of Caco-2 cells after addition of 0.3 nM toxin A mixed with increasing concentrations of SCcol, SCrec, or SCdg (A). Graph showing the quantities of toxin A bound to the cell when mixed with increasing concentration of SCcol, SCrec, or SCdg (B). Positive control with specific IgA are shown in black.

Observations:

SC prevents the association of toxin A with epithelial cells. The protection observed is due to the sugar residues present on SC.

Results

How does SC protects cells from toxin A ?

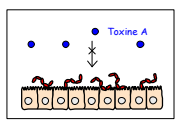
Toxine A

Hypothesis A:

SC binds to the cells and thus limits the access of the toxin: the shielding effect

When SC is preincubated with the cell before adjunction of toxin A, no significant protection is observed.

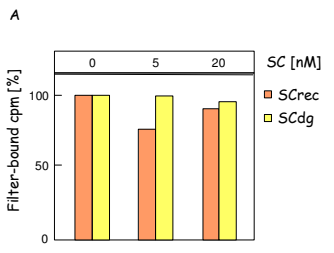
A
Filter-bound cpm [%]
SC [nM]
SCrec
SCdg

B

Recovery of SC after 60 min of incubation with the cells

	SCrec	SCdg
supernatant	99%	96%
1st washing	1%	1.4%
2nd washing	BD	BD
cell lysate	BD	BD

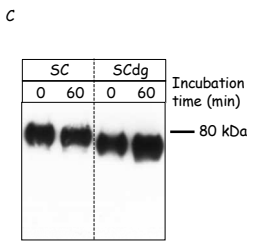
C
SC
SCdg
Incubation time (min)
80 kDa

Polarized epithelial cells were incubated with SCrec or SCdg, washed and exposed to radiolabeled toxin A. Bound toxin A was quantify by liquid scintillation counting (A). To test a possible interaction between SC and cells, SCrec or SCdg were incubated with epithelial cells for 60 min. Supernatant, washings and cell lysate were collected for subsequent determination of their SC content by ELISA. BD: below detection (B). Western Blot analysis of the integrity of SC after incubation on the cells (C).

Conclusions:

The integrity of SC is preserved, yet it does not interact with the cell monolayer in this *in vitro* model. The protection observed is not mediated through a shielding effect.

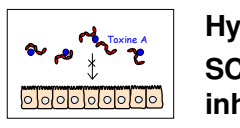
Toxine A

Hypothesis B:

SC interacts directly with the toxin, thus inhibiting its attachment to the cell.

SC does interact directly with toxin A, and thus confers the protection. The interaction is mediated through sugar residues.

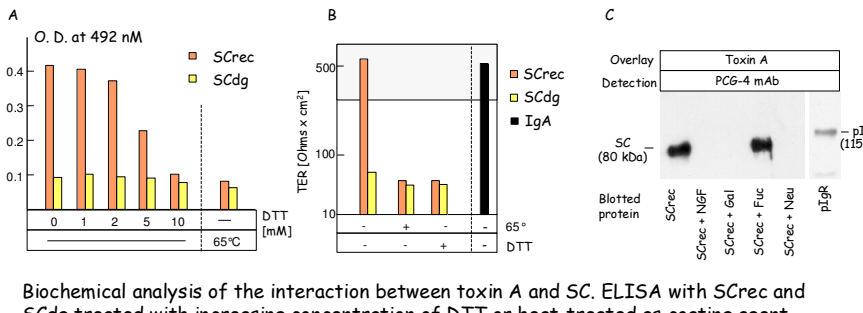
A
O. D. at 492 nM
SCrec
SCdg
DTT [mM]
65°C
B
TER [Ohms x cm²]
SCrec
SCdg
IgA
65°
DTT
C
Overlay
Detection
Toxin A
PCG-4 mAb
SC (80 kDa)
pIgR (115 kDa)
Blotted protein
SCrec
SCrec + NGF
SCrec + Gal
SCrec + Fuc
SCrec + Neu
pIgR

Biochemical analysis of the interaction between toxin A and SC. ELISA with SCrec and SCdg treated with increasing concentration of DTT or heat-treated as coating agent. After incubation of toxin A, detection was performed with anti-toxin A antibody (A). Measure of TER after 3 hours incubation of toxin A preincubated with SCrec, or SCdg untreated, treated with DTT or heat-treated (B). Overlay assay with differentially deglycosylated SC run on a 10% SDS-PAGE an incubated with toxin A (C).

Conclusions:

SC interacts directly with toxin A through its carbohydrate residues, more precisely with sialic acid and galactose. The conformation of SC polypeptide is also important for the binding properties of associated carbohydrates, as heat- or DTT-treated SC does not confer protection.

Does SC act as microbial scavenger with another pathogenic agent?

To test this issue, another bacterial protein was tested, namely the Intimin from enteropathogenic *Escherichia coli.*

As for the *Clostridium difficile* toxin A, Intimin does bind directly to SC, but not to its deglycosylated form.

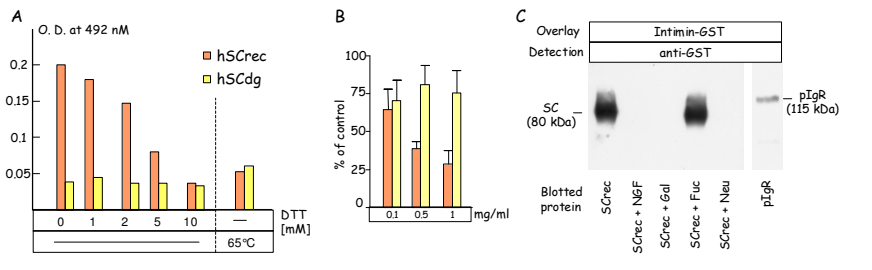
A
O. D. at 492 nM
hSCrec
hSCdg
DTT [mM]
65°C
B
% of control
mg/ml
C
Overlay
Detection
Intimin-GST
anti-GST
SC (80 kDa)
pIgR (115 kDa)
Blotted protein
SCrec
SCrec + NGF
SCrec + Gal
SCrec + Fuc
SCrec + Neu
pIgR

Biochemical analysis of the interaction between Intimin and SC. ELISA with SCrec and SCdg treated with increasing concentration of DTT or heat-treated as coating agent. After incubation of Intimin-GST, bound molecules were detected with an anti-GST antibody (A). Adhesion / effacement assay demonstrating the protective effect of SC on Hep-2 cells (B). Overlay assay with differentially deglycosylated SC run on a 10% SDS-PAGE and incubated with Intimin (C).

Conclusions:

SC interacts with Intimin through its carbohydrate residues, more precisely with sialic acid residues and galactose. The conformation of SC polypeptide is important for the binding properties of associated sugars, as heat- or DTT-treated SC abolishes the interaction.

In this particular example, SC is confirmed as a non-specific microbial scavenger.

Results

Does SC act as scavenger with a viral pathogen?

Three different strains of rotaviruses were used to study this question: the human SA11, the bovine RF, and the rhesus RRV. Rotaviruses are known to be neutralized by different carbohydrates and interact with target cells through several glycoreceptors.
However, no interaction was observed between purified rotavirus and SC, fully glycosylated or not.

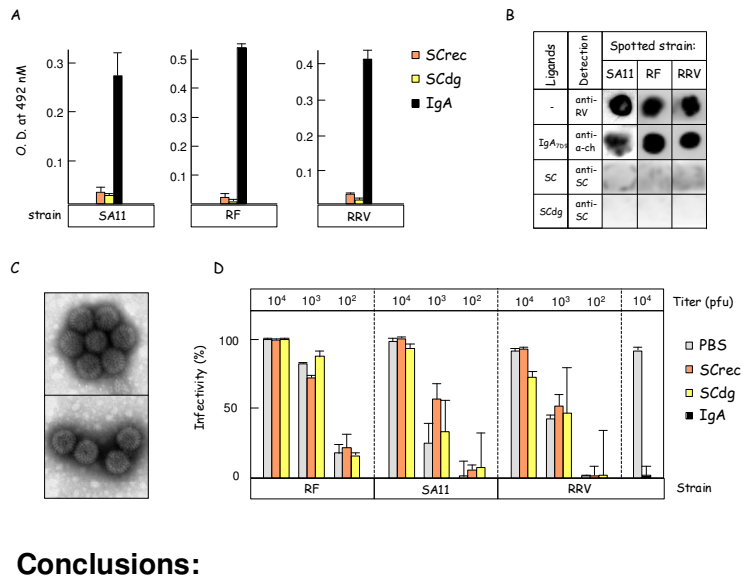
A
O. D. at 492 nM
strain
SA11
RF
RRV
SCrec
SCdg
IgA
B
Ligands
Detection
Spotted strain:
SA11
RF
RRV
anti-RV
anti-α-ch
anti-SC
SC
SCdg
C
D
Titer (pfu)
Infectivity (%)
PBS
SCrec
SCdg
IgA
Strain

The possible interaction between purified rotavirus and SC was studied with four different methods. ELISA was performed with purified virus as coating agent and incubated with SCrec or SCdg. Positive controls using specific IgA are shown in black (A). Overlay assay was performed where purified viruses were blotted and incubated with SCrec, SCdg and IgA as positive control (B). Electron microscopy pictures of rotavirus SA11 alone (up) and rotavirus SA11 mixed with SC (down) (C). Infectivity assays were performed in absence or presence of SCrec or SCdg with different titers of virus on MA104 cells (D).

Conclusions:

The three different strains of rotaviruses tested do not interact with SC. SC can therefore not be shown as a potential inhibitor of rotavirus infection.

Conclusions

The primary role of SC is the transport of secretory IgA to mucosal surfaces. It confers to the complex a remarkable stability and resistance to proteases.

Besides this, we show here that SC can interact with bacterial toxins and limit their cytotoxic effects on epithelial cells. The interaction is mediated through the numerous sugars residues present on the protein. It appears here that sialic acid and galactose play a pivotal role in these interactions.

Even if no interaction takes place between SC and rotaviruses, SC can be seen as a non specific microbial scavenger and thus complete the process of immune exclusion mainly relying on secretory IgA.

Acknowledgments

C. Perrier is supported by a grant from the Nestlé Ph.D. Program.

Nestlé Good Food, Good Life

For further information:

Clementine.perrier@chuv.hospvd.ch or Blaise.corthesy@hospvd.ch
Web site: www.immunologyresearch.ch