Prospective monitoring of a CD4$^+$ CD25^{high} CD45RO$^+$ IL-7Rα^{high} T cell population after first kidney transplantation following thymoglobulin or basiliximab induction

Vallotton Laure [1], Hadaya Karine [2], Buhler Leo [2], Ciuffreda Donatella [1], Venetz Jean-Pierre [3], Pantaleo Giuseppe [1], Pascual Manuel [3]

[1] Laboratory of AIDS Immunopathogenesis, Division of Immunology and Allergy, Department of Medicine, Centre Hospitalier Universitaire Vaudois (CHUV), Lausanne, Switzerland
[2] Service of Nephrology and Transplantation, Hôpitaux Universitaires de Genève (HUG), Geneva, Switzerland
[3] Transplantation Center, Centre Hospitalier Universitaire Vaudois (CHUV), Lausanne, Switzerland

INTRODUCTION

Our recent data (1) have demonstrated that a population of **circulating CD4$^+$ CD25^{high} T lymphocytes** (T_{act}), phenotypically characterized by the expression of **CD45RO** and **IL-7Rα** (interleukin (IL-) 7 receptor α-chain), is significantly expanded in stable liver and kidney transplant recipients. These T cells secrete **pro-inflammatory cytokines** such as **IFNγ** and **TNFα** and infiltrate the kidney allografts of patients with documented chronic humoral rejection, thus representing **alloreactive T cells**. Recently, we could show that the presence of this T cell population correlates with the clinical status of kidney transplant recipients (see Poster 246). Our conclusion is that monitoring the percentage of this circulating T_{act} population may represent a valuable tool to monitor CD4 T cell immune responses after kidney transplantation.

An important issue in kidney transplantation is the use of various **induction therapies**. Among others, **thymoglobulin** (Thymoglobuline®, Genzyme) and **basiliximab** (Simulect®, Novartis) are currently widely used; however, there are no available data yet on their impact on this alloreactive T cell population.

Thymoglobulin is a purified polyclonal anti-lymphocyte preparation obtained by immunization of rabbits with human thymocytes; it contains cytotoxic antibodies directed against antigens expressed on human T lymphocytes (CD2, CD3, CD4, CD8, CD11a, CD18, CD25, CD44, CD45, HLA-DR, HLA class I and α_2-microglobulin), and results in depletion of T lymphocytes. Possible mechanisms by which thymoglobulin may induce immunosuppression *in vivo* include: T cell clearance from the circulation and modulation of T cell activation, homing, and cytotoxic activities.

Basiliximab is a chimeric monoclonal antibody that specifically binds to and blocks the interleukin-2 receptor α-chain (IL-2Rα, CD25), which is expressed on the surface of activated as well as regulatory T cells. This specific high affinity binding to IL-2Rα competitively inhibits IL-2-mediated activation of lymphocytes, a critical pathway in the cellular immune response involved in allograft rejection.

OBJECTIVE

Prospective analysis of the CD4$^+$ CD25^{high} CD45RO$^+$ IL-7Rα^{high} activated T cell population (T_{act}) in kidney transplant recipients following either **thymoglobulin** or **basiliximab** induction.

METHODS

Flow cytometric assessment of T cell subpopulations frequency using anti-CD4, CD25, CD45RO, IL-7Rα and FoxP3 monoclonal antibodies, on peripheral blood mononuclear cells.

PATIENTS

- 32 kidney transplant recipients were studied, before and 3, 6 and 12 months after transplantation :
 - **group "THYMO"** (n=17) : induction therapy by thymoglobulin (1.5 mg/kg/day) given at days 0 and 4 and steroids (tapered in 4 days), maintenance immunosuppressive therapy by tacrolimus (FK) and mycophenolate mofetil (MMF) ;
 - **group "BSX"** (n=15) : induction therapy by basiliximab (20 mg at days 0 and 4), maintenance immunosuppressive therapy by FK, MMF and steroids.
- 27 healthy controls

RESULTS

Patients clinical outcome and demographic data

The patients clinical outcome is summarized in [**Table 1**]. All patients received their first kidney transplantation from a live related or unrelated donor. The outcome in terms of patients and graft survival was comparable in both groups. The rate of acute rejection in the first 6 months after transplantation was slightly higher in the BSX group, whilst the the serum creatinine was higher in the THYMO group.

Evolution of the T_{act} population

The evolution of the T_{act} population is depicted in [**Fig. 1**]. Overall, the T_{act} population was found to significantly increase as soon as 3 months after transplantation in both study groups; this increase was higher in the BSX group (mean : 19.39 ± 1.70%) as compared to the THYMO group (13.80 ± 1.33% ; P<0.01). At 6 months post-transplantation, the T_{act} population remained elevated in both groups as compared to healthy controls and to pre-transplantation values, but this time the percentage was comparable in both groups (BSX group : 12.29 ± 1.88% ; THYMO group : 15.12 ± 2.45% ; P=0.21). At 12 months after transplantation, the T_{act} population was still increased in a comparable extent in both groups (BSX group : 10.88 ± 0.66% ; THYMO group : 13.85 ± 1.57% ; P=0.24).

[**Table. 1**]

		Kidney transplant recipients – INDUCTION: THYMO	Kidney transplant recipients – INDUCTION: BSX	Healthy controls
		n = 17	n = 15	n = 27
Patient and graft survival (%)	at 3 months	16/16	15/15	na
	at 6 months	14/14	10/10	
	at 1 year	11/11	2/2	
Acute rejection (%)	at 6 months	1/14	2/10	na
Serum creatinine (µmol/l)	before tx	612.1	571.0	86.4
	at 3 months	137.6	114.0	
	at 6 months	129.6	122.1	
	at 1 year	145.0	127.5	
Age (years)		41.2	48.4	51.7

[**Fig. 1**]

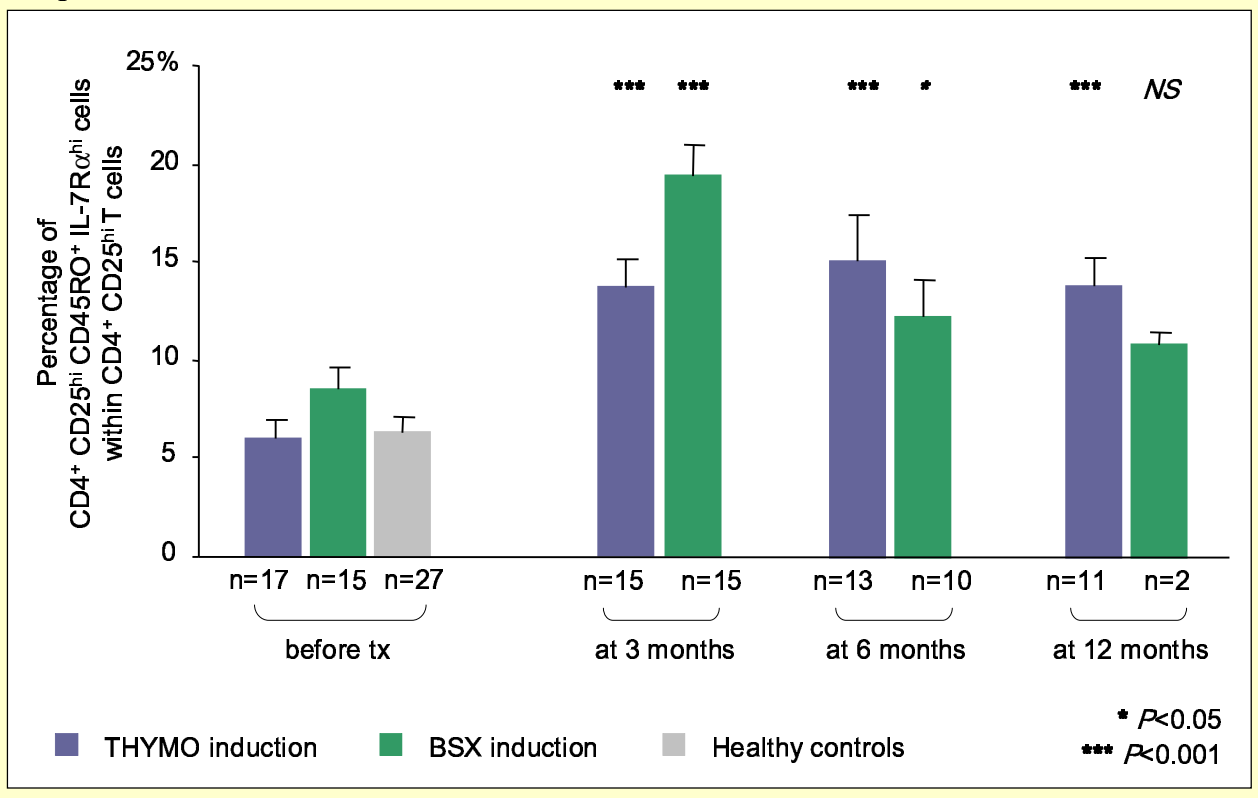
Percentage of CD4+ CD25hi CD45RO+ IL-7Rαhi cells within CD4+ CD25hi T cells
25%
20
15
10
5
0
*** *** *** * *** NS
n=17 n=15 n=27
n=15 n=15
n=13 n=10
n=11 n=2
before tx
at 3 months
at 6 months
at 12 months
THYMO induction
BSX induction
Healthy controls
* P<0.05
*** P<0.001

CONCLUSION

After kidney transplantation, a circulating CD4$^+$ CD25^{high} T cell population characterized by the expression of CD45RO and IL-7Rα expands rapidly in most recipients. The use of thymoglobulin as induction therapy may be beneficial (in terms of expansion of the T_{act} population) as compared to basiliximab early after transplantation (within the first three months). However, this difference was not significant anymore in the later timepoints.

Our results need to be confirmed by increasing the number of patients. We will also study the specific anti-donor proliferative capacity of the T_{act} population and the suppressive capacity of regulatory T cells, which are thought to be important for the outcome of transplantation.

REFERENCES

1) Codarri L, Vallotton L, Ciuffreda D, Venetz JP, Garcia M, Hadaya K, Buhler L, Rotman S, Pascual M, and Pantaleo G. Expansion and tissue infiltration of an allospecific CD4$^+$ CD25$^+$ CD45RO$^+$ IL-7Rα^{high} cell population in solid organ transplant recipients. *J Exp Med.* 204 : 1533-1541 (2007)

SUPPORTED BY a Swiss National Foundation grant (FN 323500-111275) to L. Vallotton in the context of the Swiss MD-PhD program, and by Genzyme GmbH.